Src Inhibitors with Sub nM VEGFR2 Activity – Discovery and SAR

Elena Dneprovskaia[1], Kathy Barrett[1], Jianguo Cao[1], Richard M. Fine[2], Colleen Gritzen[1], John Hood[1], Xinshan Kang[2], Boris Klebansky[2], Dan Lohse[1], Chi Ching Mak[1], Andrew McPherson[1], Glenn Noronha[1], Ved P. Pathak[1], Joel Renick[1], Richard Soll[1], Binqi Zeng[1].

[1]TargeGen, Inc., 9380 Judicial Drive, San Diego, CA 92121; [2] Discovery Partners International, Inc. and BioPredict, Inc.

Introduction

- Src is the prototype member of the Src family of cytoplasmic tyrosine kinases. Src modulates intracellular signal transduction through multiple pathways that are heavily implicated in a number of diseases such as cancer, myocardial infarction, osteoporosis, stroke and neurodegeneration.
- VEGFR2 is a member of the receptor tyrosine kinases that carry out the transmission of biochemical signals across the plasma membrane of cells. VEGFR2, which is expressed only on endothelial cells, binds the angiogenic growth factor VEGF and mediates the subsequent signal transduction through activation of its intracellular kinase activity.
- Both Src and VEGFR2 are therapeutic targets for reducing abnormal angiogenesis, proliferation and pathologically increased vascular permeability.
- Using structure-based drug design TargeGen has developed and optimized potent Src inhibitors. The crystal structure of an exemplary compound co-crystallized with Src was obtained to validate the mode of binding and to confirm interactions with the protein crucial to the activity (see MEDI 109).
- Further optimization efforts, focused on the hydrophobic pocket of Src, resulted in a new series of compounds, whose binding mode is reminiscent of the binding mode of Gleevec in Abl. In this binding mode, an apparent induced conformational change involving the αC helix and the Asp-Phe-Gly (DFG) activation loop results in the extension of the hydrophobic pocket that can accommodate a variety of groups (see MEDI 108).
- This discovery was exploited to obtain compounds that inhibit both Src and VEGFR2 in low nM ranges.
- Here TargeGen presents the discovery, optimization, and SAR of the dual Src/VEGFR2 inhibitors with low to sub nM activity.

Figure 1. Evolution of Dual Src/VEGFR2 Inhibitors Based on the P4' Region

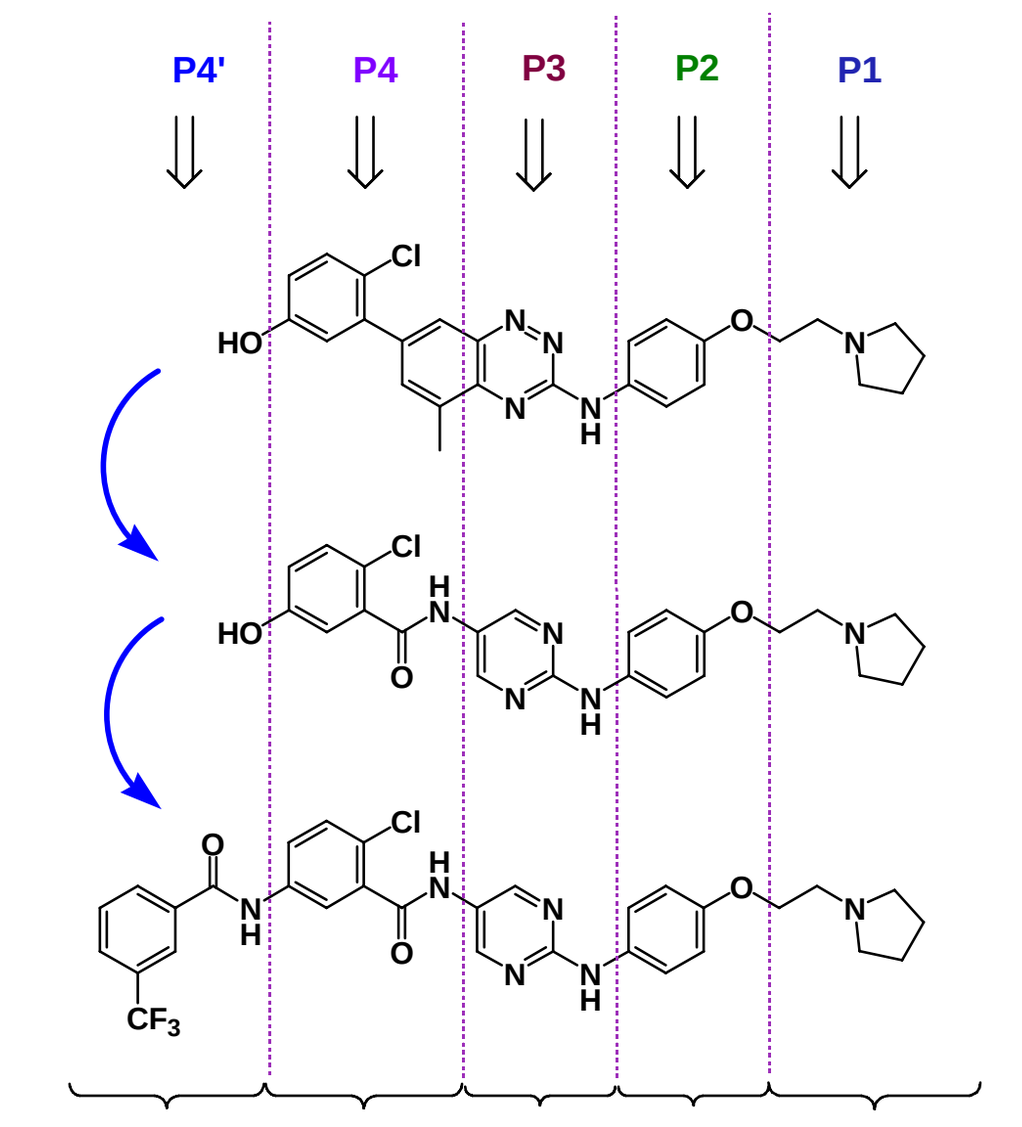

P4'
P4
P3
P2
P1
Hydrophobic pocket extension region
Hydrophobic pocket
Hinge region
Hydrophobic region
Solvent exposed region

- Based on the interactions with the protein, five distinctive regions were identified for further optimization.

Figure 2. Molecular Modeling of Compound 1 in VEGFR2 Crystal Structure 1YWN

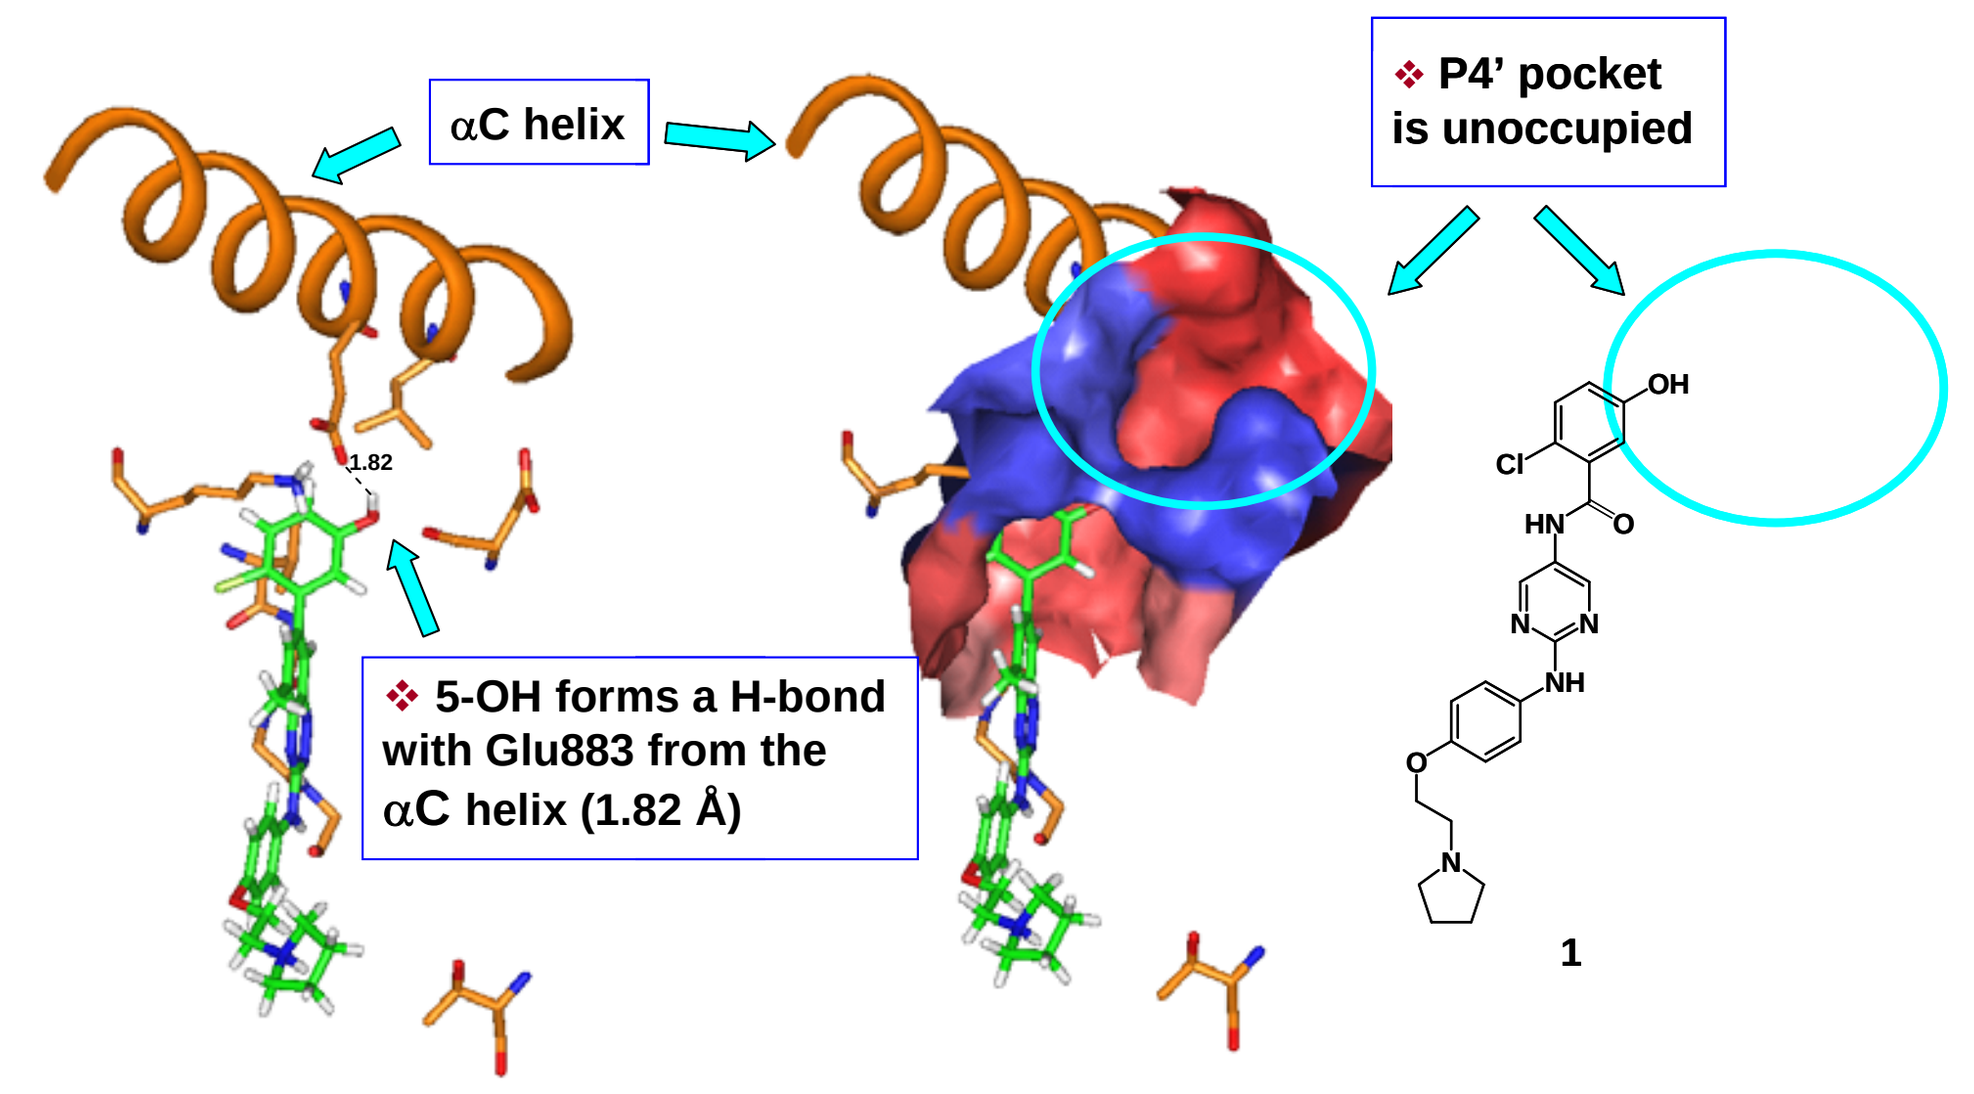

αC helix
❖ P4' pocket is unoccupied
1.82
❖ 5-OH forms a H-bond with Glu883 from the αC helix (1.82 Å)
1

Table 1. VEGFR2 Kinase Inhibition with Various Groups in the P4' Region

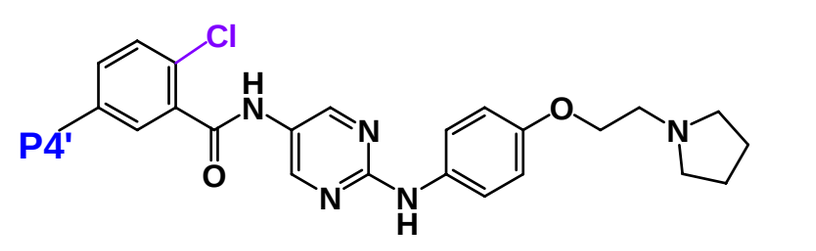

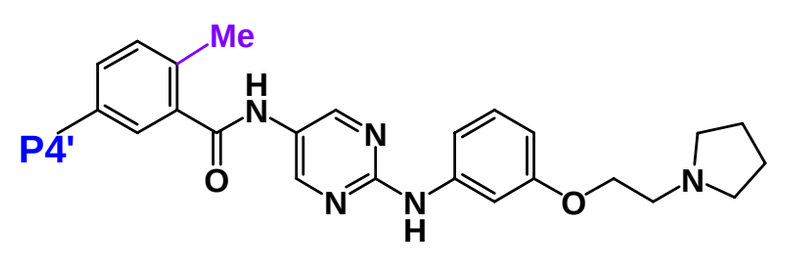

Compounds	P4'	VEGFR2 IC_{50}, nM
2		0.8
3		40.7
4		11.3
5		12.0

Compounds	P4'	VEGFR2 IC_{50}, nM
6		13.8
7		35.8
8		3347

- A hydrophobic aromatic group attached to the amide group is preferred in the P4' region (6 vs 7).
- A substitution of the phenyl ring with a 5-methyl-isoxazole ring results in a 14X loss of activity probably due to a skewed orientation of the methyl group filling the hydrophobic pocket (2 vs 4).
- A CF_3 group is preferred over smaller groups, such as F (2 vs 3).
- A CF_3 group in the meta position results in 15X better activity than CF_3 group in the para position (2 vs 5).
- 1-Amino-isoquinoline as an amide surrogate results in a loss of activity, probably due to unfavorable constraint of the P4' group, which causes steric hindrance with the protein (8 vs 6).
- 3-CF_3-phenyl is preferred in the P4' region for the VEGFR2 activity.

Table 2. Exploration of the Direction of the P4' Pocket

Compounds	P4' … P1	Src IC_{50}, nM	VEGFR2 IC_{50}, nM
9		2.1	2.7
10		65.7	621

- As seen from the modeling studies in Figure 2, the P4' group extending into the hydrophobic pocket should prefer to be in para position to the Cl group, as in compound 9. This observation was confirmed by the data from compound 10, where the P4' group in the meta position to Cl results in a 230X loss of activity. The same preference is observed for Src, where the change from para to meta substitution of the P4' group results in a 31X loss of activity.

Table 3. Chloro vs Methyl vs Hydrogen in the P4 Region

Compounds	Structure	VEGFR2 IC_{50}, nM
11		2.9
12		8.9
13		14.2

- Slight Cl > H > Me trend is observed for VEGFR2 activity for substituents on the phenyl ring in the P4 region.

Table 4. VEGFR2 Kinase Inhibition - Amide vs Urea in the P4' Region

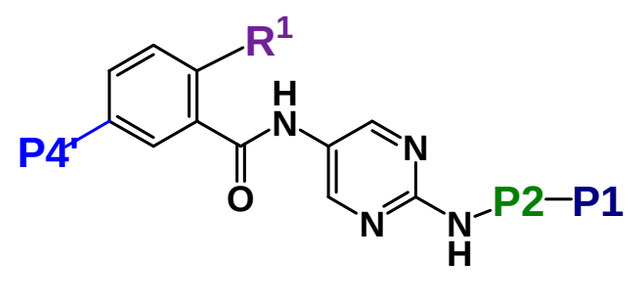

	P4'	R^1	P2-P1	VEGFR2 IC_{50}, nM
14		Me		3.2
15		Me		168
6		Me		13.8
16		Me		73.1

	P4'	R^1	P2-P1	VEGFR2 IC_{50}, nM
2		Cl		0.8
17		Cl		7.1
18		Cl		5.4
19		Cl		20.1

- Substitution of the amide group with the urea group results in a 5-6X loss of activity (6 vs 16, 2 vs 17, 18 vs 19).
- Substitution in the P4 region with the 2-chloro group results in 4-5X better activity compared to the methyl group (14 vs 2).

Table 5. Effects of P1 and P2 Groups on Activity

Compounds	P2 P1	Src IC_{50}, nM	VEGFR2 IC_{50}, nM
20		72	
21		>10000	
18		4.7	5.4
22		4.5	9.6
2		1.2	0.8

SAR of the P2-P1 Region (Table 5)

- The addition of the phenyl group in the P2 region results in a 15X gain in activity of Src, possibly due to favorable hydrophobic van der Waals interaction with the close-by lipophilic residues, such as Leu273 in Src (20 vs 21 vs 18). Analogous to Src, a similar residue Leu838 is observed in VEGFR2.
- Substituting the pyridyl ring with the phenyl results in similar activities for both Src and VEGFR2 (18 vs 22).
- A 4X increase in activity of Src is obtained with the addition of a P1 group (solvent exposed region) due to gaining an additional interaction with Asp348 (22 vs 2).
- A 12X increase in activity of VEGFR2 is obtained with the addition of a P1 group, which is also probably due to gaining an additional interaction with the protein (22 vs 2).

Table 6. Src and VEGFR2 Kinase Inhibition with Various Groups in the P2-P1 Region

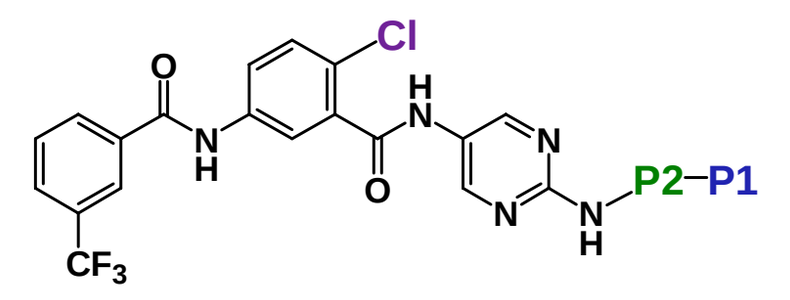

Compounds	P2-P1	Src IC_{50}, nM	VEGFR2 IC_{50}, nM
2		1.2	0.8
9		2.1	2.7
23		2.6	1.4
24		1.7	2.3
25		1.8	4.5
26		1.4	2.2

Compounds	P2-P1	Src IC_{50}, nM	VEGFR2 IC_{50}, nM
11		4.5	2.9
27		1.4	2.9
28		4.0	2.1
18		4.7	5.4
29		1.9	3.1
30		5.0	1.2

- Both Src and VEGFR2 activity are improved by addition of a P1 group due to gaining an additional interaction with the protein. However, all the linkers give similar activity provided that they are 4-5 atoms in length to make the interaction.
- There is an observed slight preference for the ether linker for the VEGFR2 activity. All other linkers are similar in activity, such as ether > CH2 linker ~ amide ~ sulfonamide (11 and 27, 24 and 25, 26 and 27).
- In case of the sulfonamide linker, para and meta substitution gives similar activities for VEGFR2 (27 and 28).
- In case of the ether linker para substitution results in a 3x gain in VEGFR2 activity compared to meta substitution, and a 2X gain in Src activity (2 vs 9).
- Substitution of the phenyl ring with the pyridyl ring in the P2 region gives similar activities (26 vs 30) for VEGFR2 (as also seen in the case of 18 vs 22), with slight preference for the phenyl group in the case of Src.

Table 7. Kinase Profile of Compound 2 with Selected Kinases

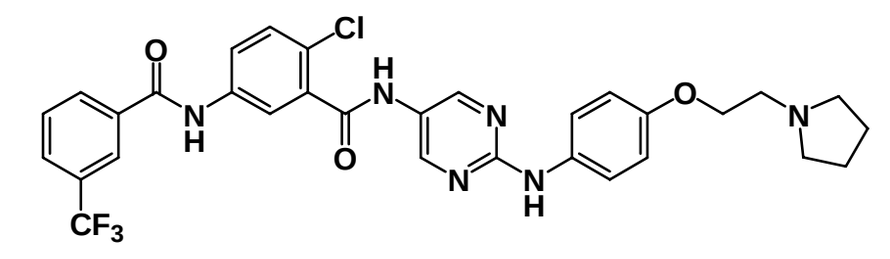

2

Src IC_{50}, nM	VEGFR2 IC_{50}, nM	Yes IC_{50}, nM	Lck IC_{50}, nM	Fyn IC_{50}, nM	PDGFRβ IC_{50}, nM	EGFR IC_{50}, nM	Abl IC_{50}, nM
1.2	0.8	0.46	2.2	3.6	0.67	97.5	1.8

- Compound 2 inhibits Src family kinases and growth factor receptors, such as VEGFR2, PDGFRβ, and EGFR. It also inhibits Abl, which is closely related in the kinase domain to Src.

Figure 3. Molecular Modeling of Compound 2 in VEGFR2 Crystal Structure 1YWN

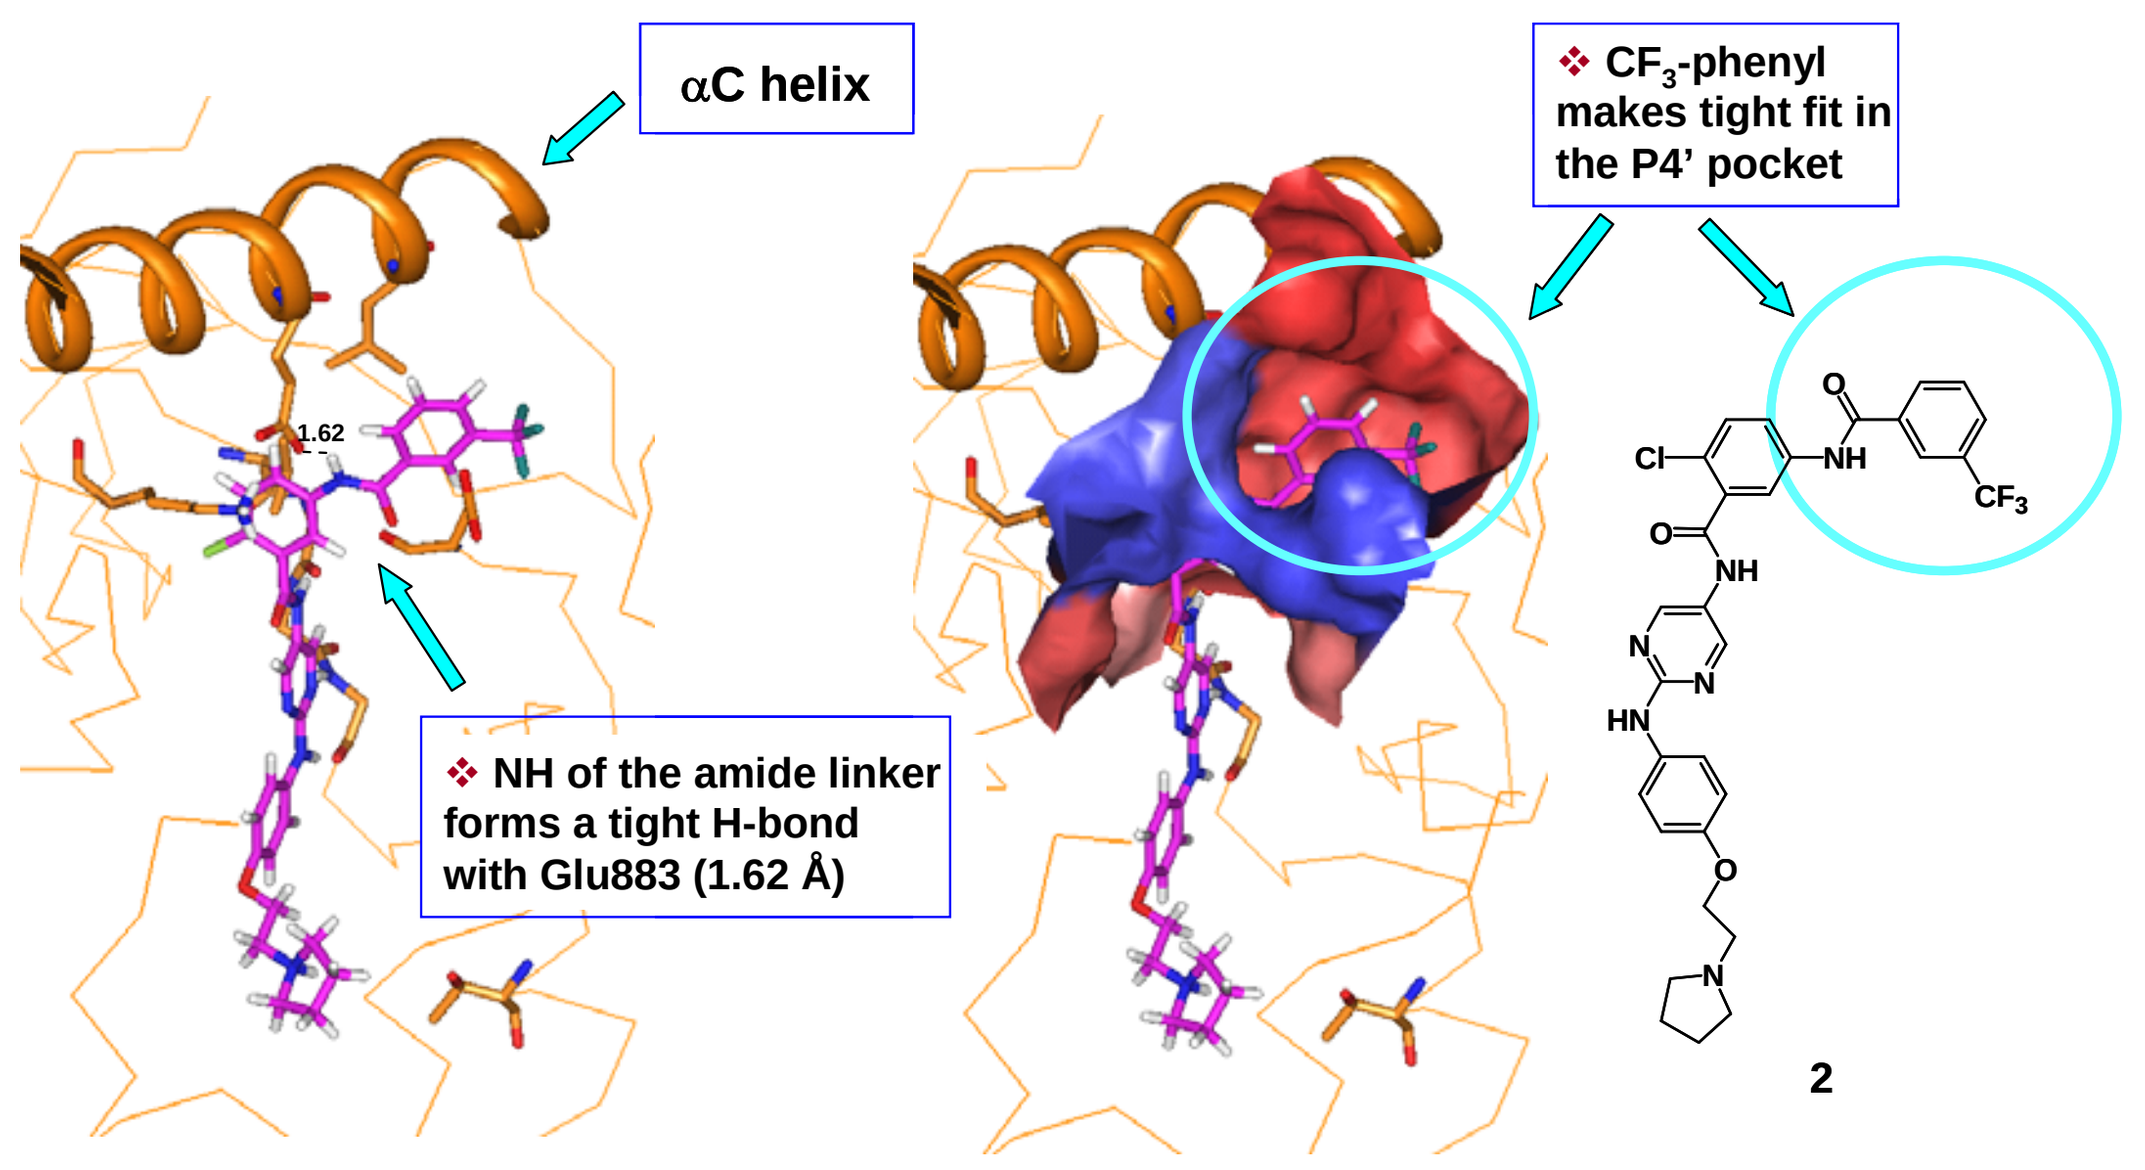

αC helix
❖ CF_3-phenyl makes tight fit in the P4' pocket
1.62
❖ NH of the amide linker forms a tight H-bond with Glu883 (1.62 Å)
2

Figure 4. Molecular Modeling of Compound 2 in VEGFR2 Crystal Structure 1YWN

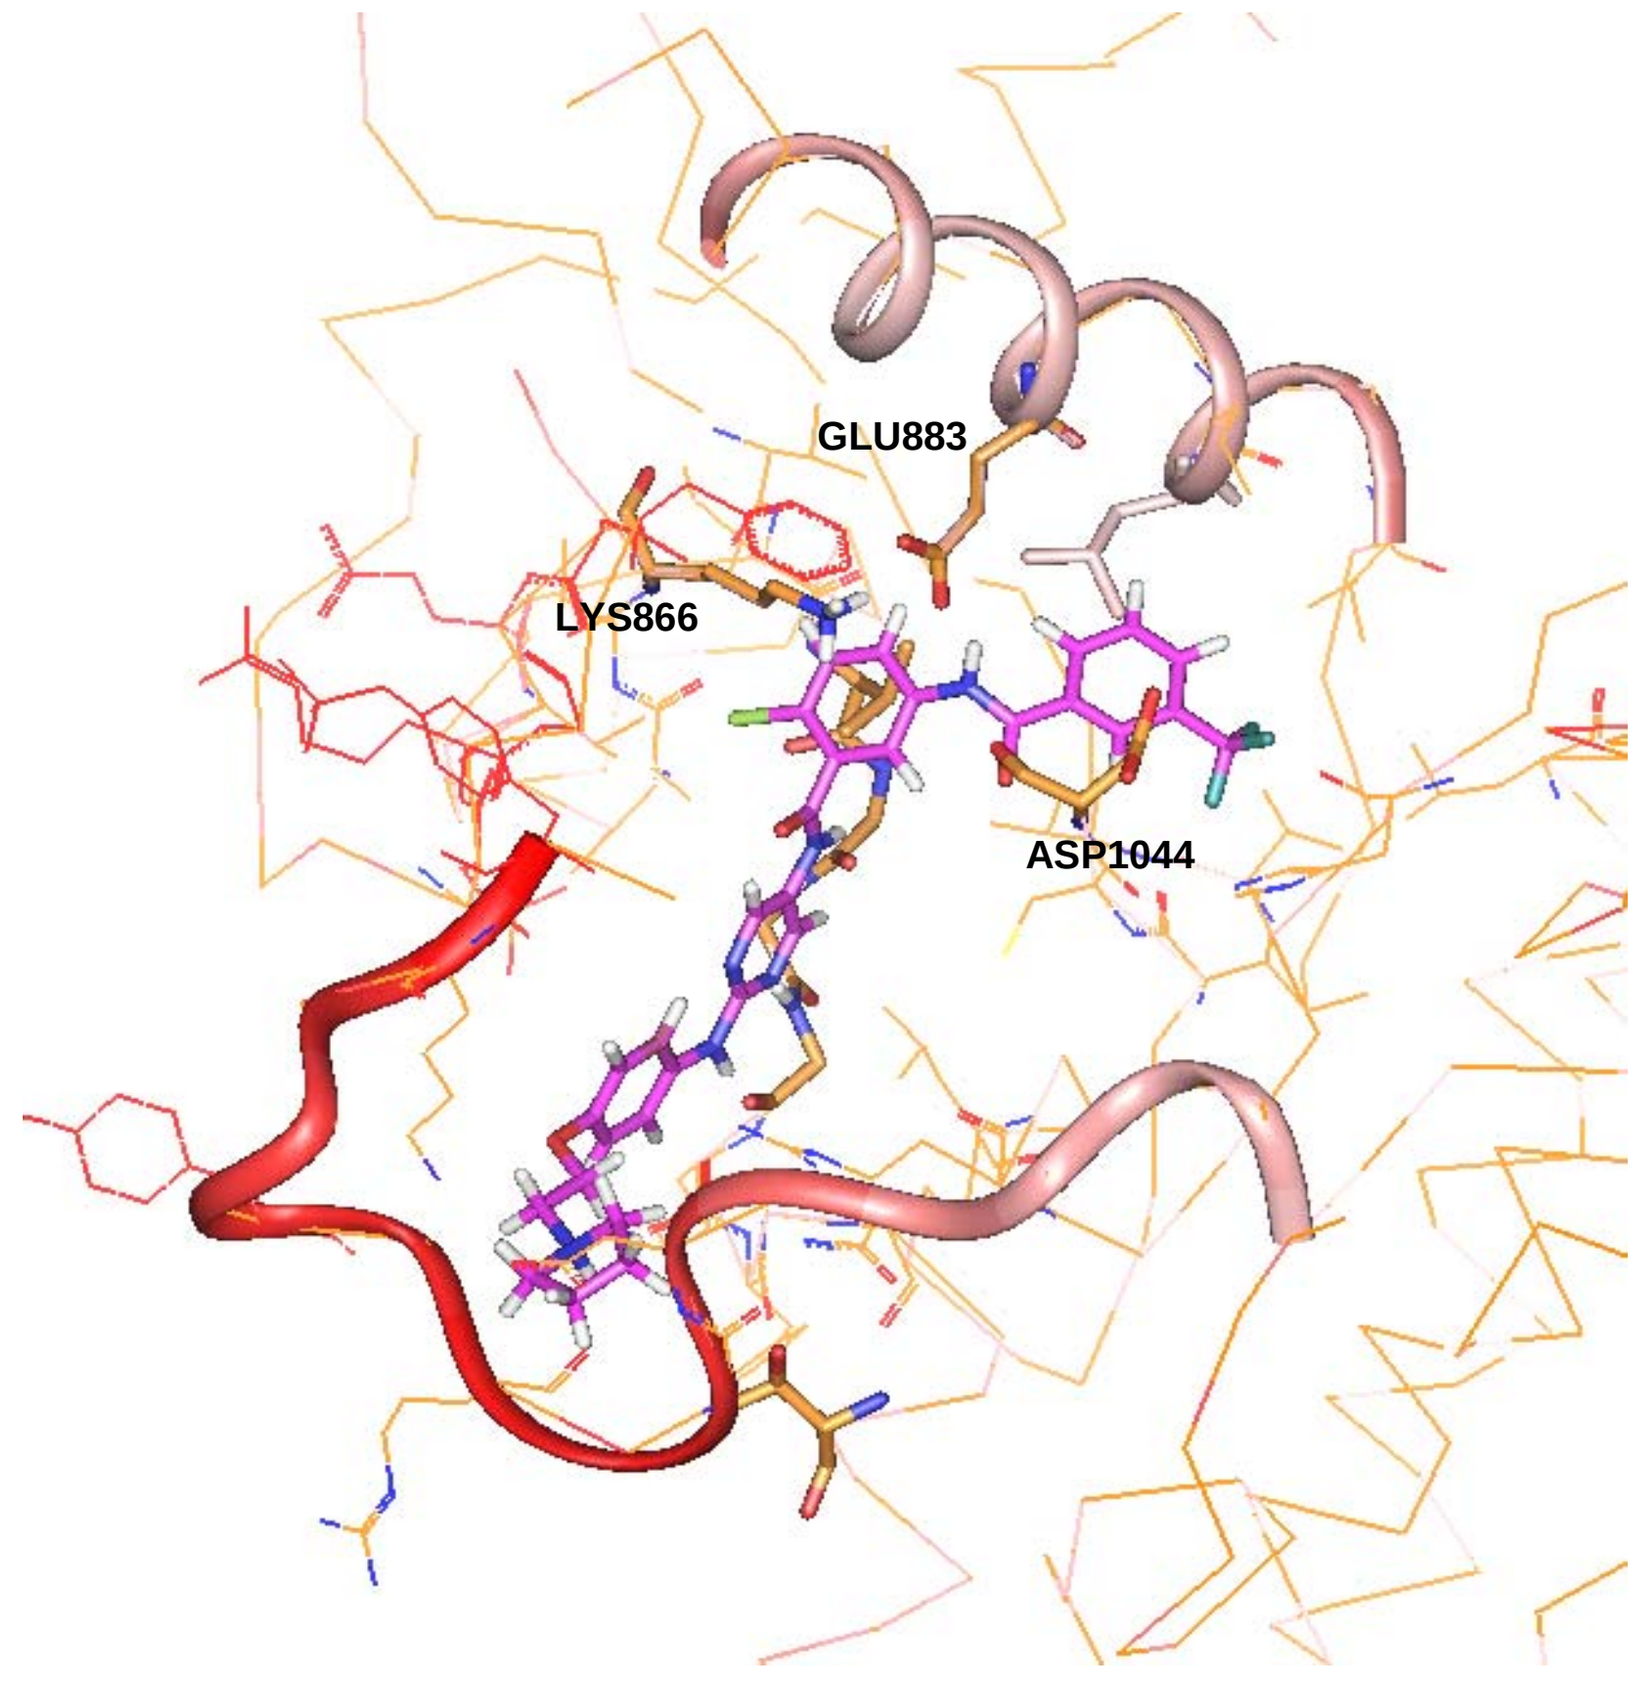

GLU883
LYS866
ASP1044

- The nitrogen and NH of the pyrimidine ring make H-bond interactions with the Cys917 residue of the hinge region.
- The NH of the amide group in the P4' region makes an interaction with Glu883.
- The phenyl group in the P4 region possibly makes hydrophobic interactions with Lys886 and Val914.
- The extended hydrophobic pocket contains the Leu887, Asp1044 and Glu883 residues.
- Formation of the P4' pocket in VEGFR2 apparently involves the movement of the αC helix and the complete translocation of the DFG activation loop away from the activation site. In VEGFR2 this loop is extremely flexible, even in the activated form of the protein, and in most cases is not seen in the crystal structures.

Figure 5. Comparison of the Active Src without P4' Pocket (Green) with the Active Src with P4' Pocket (Blue) with Compound 2

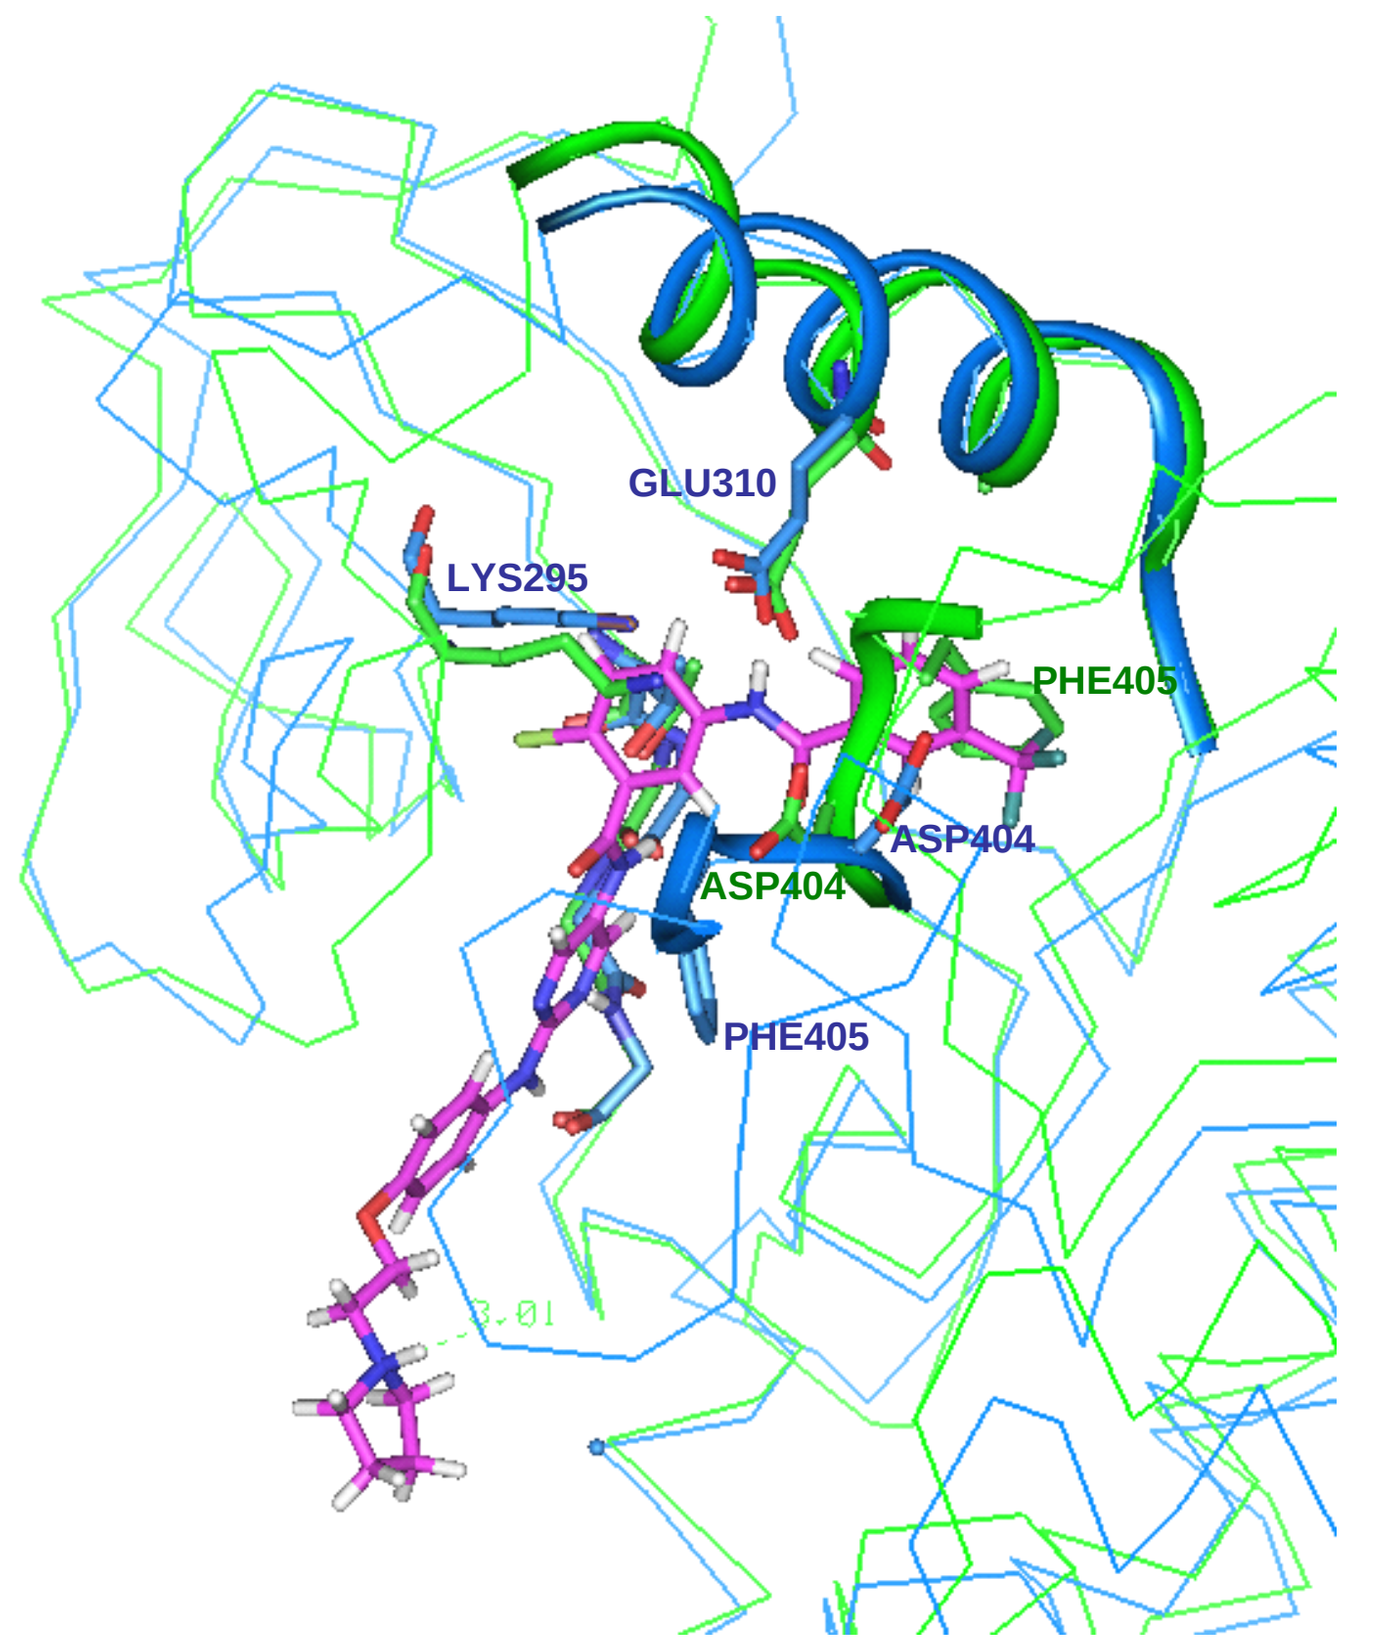

GLU310
LYS295
PHE405
ASP404
ASP404
PHE405

- Upon activation of Src, the αC helix moves in toward the binding pocket, thus creating conflicts with the extension linkage to the P4' substitutions. A portion of the activation loop that follows the conserved DFG loop packs against the αC helix and eliminates the potential P4' pocket (see Asp404 and Phe405 location). As a result, most designs of known Src inhibitors do not extend beyond the usual themes of the hydrophobic pocket and stop at the P4 region.
- The induced conformational change resulting in the formation of the P4' pocket in active Src most likely requires two relaxations. One is the relaxation of the αC helix to avoid the bumps with the extensions in the TargeGen compounds, and the other is the relaxation of the DFG activation loop to form the pocket (see Asp404 and Phe405 location). Similar relaxation of the protein to form the extended hydrophobic pocket was reported by Amgen with a 2-aminoquinazoline analog bound to Lck (*J. Med. Chem*, 2006, doi:10.1021/jm0605482).

Highlights and Summary

- Having validated a structure-based design concept of making an interaction with Glu310 of the αC helix deep within the hydrophobic pocket of Src, Targegen has developed a novel class of Src inhibitors, that extend past the conventional hydrophobic pocket.
- These compounds utilize a binding mode in Src that arises from an induced conformational change involving the αC helix and the DFG activation loop movement.
- This discovery was exploited to obtain compounds that inhibit both Src and VEGFR2 in low nM ranges.
- Both Src and VEGFR2 have been shown to be therapeutic targets for reducing abnormal angiogenesis, proliferation and pathologically increased vascular permeability.

2006 ACS Fall National Meeting San Francisco, CA, September 9-14