Optimization of a novel Src inhibitor series to obtain sub-nM compounds

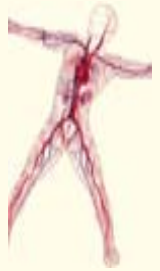

***Chi Ching Mak*[1], *Kathy Barrett*[1], *Jianguo Cao*[1], *Richard M. Fine*[2], *Colleen Gritzen*[1], *John Hood*[1], *Jason Kang*[2], *Andrew McPherson*[1], *Glenn Noronha*[1], *Ved P. Pathak*[1], *Joel Renick*[1], *Richard Soll*[1], *Ute Splittgerber*[1], *Binqi Zeng*[1]**

[1]TargeGen, Inc. 9380 Judicial Drive, San Diego, CA 92121; [2]Discovery Partners International, Inc. and BioPredict, Inc.

Introduction

Src is one of the three ubiquitously expressed Src family kinases and is the best known cellular kinase. There is significant evidence that Src is overexpressed in various human tumors and increased Src activity is observed in metastatic tumors, particularly in colon and breast tumors. Increased Src activity has also been found in VEGF-mediated increase in vascular permeability as well as in bone disorders. As a result, Src kinase has been considered a therapeutic target for cancer, osteoporosis, stroke and myocardial infarction.

TargeGen has designed and optimized a novel series of benzotriazines that are used to target Src. The focus in this poster will be the work carried out to obtain sub–nM inhibitors of Src via elaboration of the R group (**Figure 1**), using a variety of substituted and unsubstituted alkyl, aryl and heteroaryl groups. Presented herein are details for the design (for syntheses, see poster MEDI 64) of the inhibitors with the R group deep within the Src hydrophobic pocket, the resulting SAR, and the optimization that led to highly potent sub–nM Src inhibitors from this series.

Figure1: Design - template regions

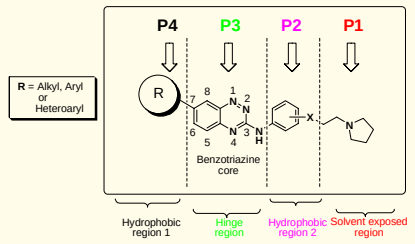
P4
P3
P2
P1
R = Alkyl, Aryl or Heteroaryl
R
Benzotriazine core
Hydrophobic region 1
Hinge region
Hydrophobic region 2
Solvent exposed region

The benzotriazine series – early SAR for Src

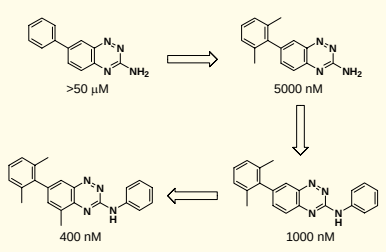
>50 μM
5000 nM
1000 nM
400 nM

Lead compound **1**

Earlier work based on **1** indicated that optimization of the P2-P1 regions (see **Figure 1**) plays a crucial role in obtaining low nM inhibition for Src (please see poster MEDI 66)

Src activity can be improved or maintained with small hydrophobic or uncharged hydrophilic substituents introduced at position 5 or 6 of the benzotriazine core (P3 region) (e.g. methyl group in **1**)

While keeping the P2-P1 section on the right hand side constant as in scaffolds **2** and **3**, modification of the R group in the P4 region as described below has led to highly potent sub-nM Src inhibitors

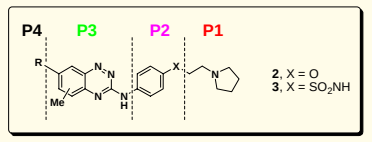
P4
P3
P2
P1
R
Me
X
2, X = O
3, X = SO_2NH

All following P4 modified compounds fall into 3 major categories based on the resulting SAR:

a) Hydrophobic aryl moieties with mono- or di- substitution (Category A)

b) Hetero-aromatic or alkyl moieties (Category B)

c) Aryl moieties with appropriate donor groups that are additionally mono-, di- or tri- substituted (Category C)

SAR of Category A compounds (Tables 1-4)

1. The aryl group in the P4 region is crucial for the activity of compound. Simple phenyl ring increases the activity to nM inhibition (compare **Table 1: 7** with **Table 2: 13** and **Table 4: 28** with **31**)
2. Compounds with or without a methyl substitution on the core exhibit similar potency (compare **5** with **9**)
3. With 2- or 2,6- substitution, Cl or Me (**15**-**17**), potency increases by at least 10 fold when compared to an unsubstituted phenyl group (**13**)
4. The P4 binding pocket is not tolerant to groups at 2-position larger in size than a Cl or Me (**29** and **30**)
5. The P4 binding pocket is less tolerant to hydrophilic groups (**32** and **33**)
6. With 4-F, 5-F or 5-CN in the aryl group, 2,4- or 2,5- disubstituted compounds maintain good activity (e.g. **4**, **6** and **25**)
7. With methoxy in 5-position (**11** and **18**), however, is not tolerated

Table 1: Enzymatic data of Category A compounds containing scaffold 2 with a 6-methyl or des-methyl core

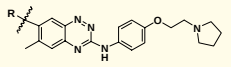

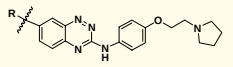

Compound	R	Src IC_{50} (nM)
4		26
5		12
6		7

Compound	R	Src IC_{50} (nM)
7	H	>10,000
8		30
9		15

- R group in P4 region is essential to the activity of the inhibitors (see **7**-**9**)
- Methyl group at C6 in the benzotriazine core is not crucial to the activity (**5** vs. **9**)

Table 2: Enzymatic data of Category A compounds containing scaffold 2 with a 5-methyl core

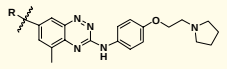

Compound	R	Src IC_{50} (nM)
10		1,200
11		778
12		505
13		341

Compound	R	Src IC_{50} (nM)
14		208
15		35
16		11
17		7

- Inhibitors with 2,6-disubstituted (Cl or Me) aryl groups, exhibit low nM activity (compare **13** with **15**-**17**)

Table 3: Enzymatic data of Category A compounds containing scaffold 3 with a 5-methyl core

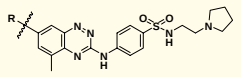

Compound	R	Src IC_{50} (nM)
18		10,000
19		10,000
20		2,900
21		631
22		353

Compound	R	Src IC_{50} (nM)
23		282
24		51
25		25
26		14
27		9

- *meta*-Substitution in aryl group is only tolerant to small group such as CN (**25**)
- Inhibitors with donor ability have moderate activity (**21**-**24**)

Table 4: Enzymatic data of Category A compounds containing scaffold 3 with a 6-methyl core

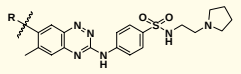

Compound	R	Src IC_{50} (nM)
28	H	>10,000
29		5,000
30		2,216
31		315

Compound	R	Src IC_{50} (nM)
32		92
33		72
34		20
35		12

- *meta*-Substitution in aryl group is only tolerant to small group (compare **30** with **35**)
- *ortho*-Substitution is not tolerant to large or hydrophilic groups (see **29**, **32** and **33**)

SAR of Category B compounds (Tables 5 and 6)

1. In general, the hydrophobic P4 region does not tolerate hetero-aromatics. Please compare the activities in the following examples: (a) **Table 5: 36** with **Table 3: 24**; (b) **Table 5: 37** or **38** with **Table 2: 13**; (c) **Table 5: 39** with **Table 2: 17**; and (d) **Table 5: 40** with **Table 1: 6**
2. The P4 region also does not tolerate alkyl groups (see **48**, **49** and **51**)
3. Of all the compounds with hetero-aromatics in P4 region, compounds **41** and **42** exhibit the best inhibitory activity at ~50 nM

Table 5: Enzymatic data of Category B compounds containing scaffold 2 with a 5- or 6- methyl core

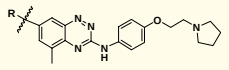

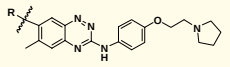

Compound	R	Src IC_{50} (nM)
36		~10,000
37		1,800
38		1,500
39		207
40		204

Compound	R	Src IC_{50} (nM)
41		61
42		44

- Not tolerant to any hetero-aromatics in P4 region

Table 6: Enzymatic data of Category B compounds containing scaffold 3 with a 5- or 6- methyl core

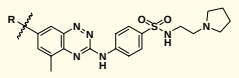

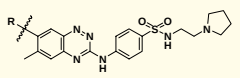

Compound	R	Src IC_{50} (nM)
43		>10,000
44		>10,000
45		10,000
46		10,000
47		144

Compound	R	Src IC_{50} (nM)
48		10,000
49		10,000
50		8,000
51		1,772
52		1,730
53		880

- Not tolerant to any hetero-aromatic or alkyl groups in P4 region

Design Strategy for Category C compounds

- The active conformation of Src presents a glutamic acid (Glu 310) from the C-helix with an unique binding opportunity deep within the hydrophobic pocket. This Glu (310) is part of the Asp-Lys-Glu triad that is a part of the Src activation mechanism.
- We decided to capitalize on this unique opportunity by making use of a donor group on the phenyl ring. Molecular modeling (**Figure 2**) predicted that a donor located on the *meta*-position would interact optimally with Glu (310).

Figure 2: Binding of compound 56 within the Src kinase domain

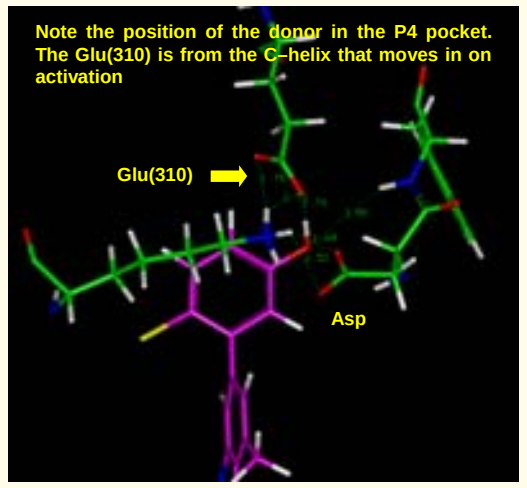
Note the position of the donor in the P4 pocket. The Glu(310) is from the C–helix that moves in on activation
Glu(310)
Asp

SAR of Category C compounds (Tables 7-9)

1. Interaction of *meta*-hydroxyl aryl group with the Glu residue within the hydrophobic pocket is extremely critical for the excellent activity of the compounds (compare **Table 2: 11** with **Table 7: 56** and see **Figure 2**)
2. Compounds with less acidic aryl group exhibit ~13-170 fold less potent inhibitory activity (compare **Table 7: 68** with amino compounds **Table 3: 21**-**24**)
3. Compounds with *ortho/para*- hydroxyl aryl group are less potent (compare **Table 7: 59** with **62** or **Table 8: 65** with **68**)

IC_{50} Value Determinations: Our compounds were tested using the KinaseGlo assay. The test compound at concentrations ranging from 1nM through 100 μM was treated with recombinant human c-Src (28 ng/well, Panvera-Invitrogen, Madison, WI), ATP (2 μM), and a tyrosine kinase substrate (PTK2, 250 μM, Promega, Corp., Madison, WI) in Src kinase reaction buffer (Upstate USA, Lake Placid, NY) at room temperature for 90 minutes. The residual ATP was determined using a luciferase-based assay (KinaseGlo, Promega Corp.) as a measure of kinase activity. The data from four wells were averaged and used to determine the IC50 using Prism software (GraphPad Software, San Diego, CA).

SAR of Category C compounds (Tables 7-9) (continued…)

4. Again, compounds with 5- or 6- methyl substituted core exhibited similar activity (compare **56** with **62**)
5. Single digit nM or sub-nM activities are obtained for compounds with a *meta*-hydroxyl aryl group, 2- or 2,6- substituted with Me, F, Cl or Br (e.g. **67**-**74**)
6. Not tolerant to linker between *meta*-hydroxy aryl group and the core (compare **75** and **77**), probably wrong orientation of the hydroxyl group

Table 7: Enzymatic data of Category C compounds containing scaffold 2 with a 5- or 6- methyl core

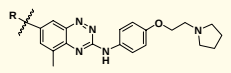

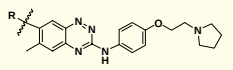

Compound	R	Src IC_{50} (nM)
54		75
55		8
56		3.5
57		1.1
58		0.9

Compound	R	Src IC_{50} (nM)
59		79
60		18
61		14
62		4.2

- *para*-OH aryl groups in the P4 region are less favorable (see **54** and **59**)
- *meta*-OH aryl groups with mono- or di- substitution in *ortho* positions provide inhibitors with single digit nM or sub-nM activity (**56**-**58**)

Table 8: Enzymatic data of Category C compounds containing scaffold 3 with a 5-methyl core

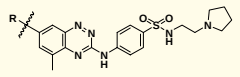

Compound	R	Src IC_{50} (nM)
63		156
64		123
65		13
66		5.2
67		4.1
68		3.6

Compound	R	Src IC_{50} (nM)
69		3.3
70		3.1
71		1.5
72		1
73		0.8
74		0.7

- *para*- or *ortho*- OH aryl groups provide less potent inhibitors (see **63** and **64**)
- *meta*-OH aryl groups with mono- or di- substitution in *ortho* positions provide inhibitors with single digit nM or sub-nM activity (**66**-**74**)

Table 9: Enzymatic data of Category C compounds containing scaffold 3 with a 6-methyl core

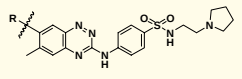

Compound	R	Src IC_{50} (nM)
75		10,000
76		8.5
77		1.6

- Not tolerant to linker in between *meta*-hydroxyl aryl group and the core

Highlights

1. A novel series of benzotriazine for Src inhibitors has been optimized from high μM to sub-nM inhibitory activity
2. Aryl moiety in the P4 region, particularly with 2- or 2,6- substitutions (Cl or Me), is essential to the activity of TargeGen benzotriazine inhibitors
3. Heteroaryl or alkyl moieties, in general, are not tolerated in the P4 region of TargeGen benzotriazine inhibitors
4. Introduction of a *meta*-donor (e.g. OH) aryl group in the P4 region significantly enhances the activity. We postulate that this arises from a hydrogen bonding interaction from the donor OH and Glu (310) deep within the hydrophobic pocket of Src P4 region
5. For compounds with a *meta*-hydroxyl aryl group, mono- or di- substitution in *ortho*-position (e.g. Me, F, Cl or Br) enhance the potency to low- or sub- nM activity
6. The design and optimization of the donor interaction with a conserved Glu residue within the hydrophobic pocket has led us to expand TargeGen oncology program to include Bcr-Abl inhibitors, in particular, targeting the T315I mutant for CML (please see poster MEDI 251)

2006 ACS Spring National Meeting, Atlanta, GA March 26 – 30